MPC-7869, a Selective Aβ42-Lowering Agent, Delays Time to Clinically Significant Psychiatric Adverse Events in Alzheimer's Disease: Analysis from a 12-Month Phase 2 Trial

Jacobo Mintzer[1], Gordon Wilcock[2], Sandra Black[3], Kenton Zavitz[4], Mark Laughlin[4], Suzanne Hendrix[4]

[1]Department of Psychiatry, Medical University of South Carolina, USA; [2]Oxford University, UK; [3]Sunnybrook & Women's, University of Toronto, Canada; [4]Myriad Pharmaceuticals, Inc., Salt Lake City, UT, USA

MPC-7869 Clinical Rationale

- Novel anti-amyloid treatment strategy for AD
- Selective Aβ42-Lowering Agent (SALA) *in vitro* & *in vivo*
 - Allosteric modulation of γ-secretase
- Reduces insoluble amyloid in mouse brain
- Improves spatial reference learning and memory performance in transgenic mice
- Effective concentrations in animal models achievable in humans at doses that have been well tolerated
- Hypothesis to be tested:
 - Subjects receiving MPC-7869 will have a delay in the onset of psychiatric adverse events
 - Hypothesis is based on the potential disease modifying effects of SALA agents

Methodology

- Randomized, Double-Blind, Placebo-Controlled Phase 2 Study
 - 31 sites in Canada and the United Kingdom
- Mild to moderate Alzheimer's (MMSE 15-26)
 - Stable cholinesterase inhibitor allowed
- 207 Subjects in 3 treatment groups (1:1:1)
 - 400 mg BID, 800 mg BID or Placebo BID
- 12 months treatment (optional 1 year follow-on study available in Canada)
- Primary Efficacy: Cognition, Activities of Daily Living, Global Function
- Safety Endpoints
 - Incidence of adverse events
 - Changes in physical examinations
 - Clinical laboratory results
- A post hoc exploratory analysis was performed comparing the number of and time to adverse psychiatric events between treatment groups

Trial Profile

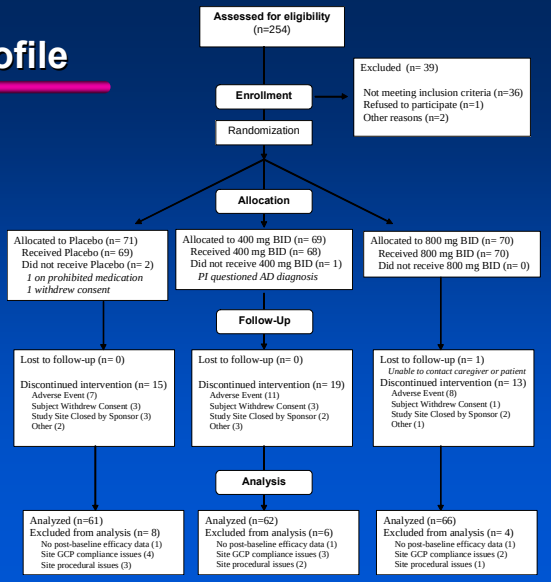
Assessed for eligibility (n=254)
Enrollment
Excluded (n= 39)
Not meeting inclusion criteria (n=36)
Refused to participate (n=1)
Other reasons (n=2)
Randomization
Allocation
Allocated to Placebo (n= 71)
Received Placebo (n= 69)
Did not receive Placebo (n= 2)
1 on prohibited medication
1 withdrew consent
Allocated to 400 mg BID (n= 69)
Received 400 mg BID (n= 68)
Did not receive 400 mg BID (n= 1)
PI questioned AD diagnosis
Allocated to 800 mg BID (n= 70)
Received 800 mg BID (n= 70)
Did not receive 800 mg BID (n= 0)
Follow-Up
Lost to follow-up (n= 0)
Discontinued intervention (n= 15)
Adverse Event (7)
Subject Withdrew Consent (3)
Study Site Closed by Sponsor (3)
Other (2)
Lost to follow-up (n= 0)
Discontinued intervention (n= 19)
Adverse Event (11)
Subject Withdrew Consent (3)
Study Site Closed by Sponsor (2)
Other (3)
Lost to follow-up (n= 1)
Unable to contact caregiver or patient
Discontinued intervention (n= 13)
Adverse Event (8)
Subject Withdrew Consent (1)
Study Site Closed by Sponsor (2)
Other (1)
Analysis
Analyzed (n=61)
Excluded from analysis (n= 8)
No post-baseline efficacy data (1)
Site GCP compliance issues (4)
Site procedural issues (3)
Analyzed (n=62)
Excluded from analysis (n=6)
No post-baseline efficacy data (1)
Site GCP compliance issues (3)
Site procedural issues (2)
Analyzed (n=66)
Excluded from analysis (n= 4)
No post-baseline efficacy data (1)
Site GCP compliance issues (2)
Site procedural issues (1)

Prospective Statistical Analysis Plan

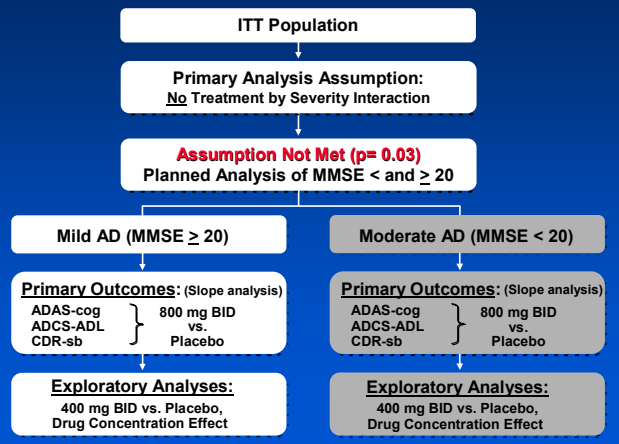
ITT Population
Primary Analysis Assumption:
No Treatment by Severity Interaction
Assumption Not Met (p= 0.03)
Planned Analysis of MMSE < and ≥ 20
Mild AD (MMSE ≥ 20)
Moderate AD (MMSE < 20)
Primary Outcomes: (Slope analysis)
ADAS-cog
ADCS-ADL
CDR-sb
800 mg BID vs. Placebo
Primary Outcomes: (Slope analysis)
ADAS-cog
ADCS-ADL
CDR-sb
800 mg BID vs. Placebo
Exploratory Analyses:
400 mg BID vs. Placebo,
Drug Concentration Effect
Exploratory Analyses:
400 mg BID vs. Placebo,
Drug Concentration Effect

Demographics by Treatment Group

Mild patients (MMSE ≥20)

	Placebo (n=46)	400 mg BID (n=36)	800 mg BID (n=48)
% of Total Patients	75%	58%	73%
Age	76	75	76
% AChEI Use	97%	94%	94%
MMSE	22.9	23.1	22.8
ADAS-cog (*80 point)	27.5	28.6	28.3
ADCS-ADL	58.7	61.4	59.8
CDR-sb	5.7	5.0	6.0

Safety: Discontinuations Over Time

Cumulative Discontinuations Over Time

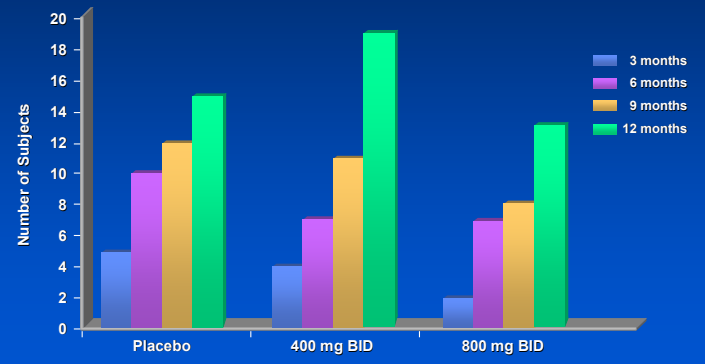
20
18
16
14
12
10
8
6
4
2
0
Number of Subjects
Placebo
400 mg BID
800 mg BID
3 months
6 months
9 months
12 months

Overall discontinuations in 12 months ~ 20%

Safety: Adverse Events That Occurred in >5% of Patients or Demonstrated Statistical Significance (p<0.10)

Preferred Term	Placebo (n=66)	400mg BID (n=71)	800mg BID (n=70)	Total (n=207)	Placebo vs 400mg BID	Placebo vs 800mg BID	400 vs 800mg BID
Diarrhea	5 (7.6)	8 (11.3)	7 (10.0)	20 (9.7)	0.565	0.765	1.000
Nausea	4 (6.1)	7 (9.9)	7 (10.0)	18 (8.7)	0.535	0.533	1.000
Dizziness	4 (6.1)	9 (12.7)	5 (7.1)	18 (8.7)	0.247	1.000	0.399
Urinary tract infection	5 (7.6)	7 (9.9)	5 (7.1)	17 (8.2)	0.766	1.000	0.764
Agitation	5 (7.6)	7 (9.9)	4 (5.7)	16 (7.7)	0.766	0.739	0.532
Confusional state	2 (3.0)	10 (14.1)	4 (5.7)	16 (7.7)	0.032	0.681	0.157
Upper respiratory tract infection	7 (10.6)	4 (5.6)	2 (2.9)	13 (6.3)	0.354	0.090	0.681
Vomiting	3 (4.5)	3 (4.2)	6 (8.6)	12 (5.8)	1.000	0.495	0.326
Headache	6 (9.1)	3 (4.2)	3 (4.3)	12 (5.8)	0.313	0.315	1.000
Constipation	3 (4.5)	4 (5.6)	4 (5.7)	11 (5.3)	1.000	1.000	1.000
Depression	4 (6.1)	1 (1.4)	6 (8.6)	11 (5.3)	0.196	0.746	0.063
Eosinophilia	0 (0)	3 (4.2)	6 (8.6)	9 (4.3)	--	0.028	
Nasopharyngitis	4 (6.1)	0 (0)	5 (7.1)	9 (4.3)	0.051	--	0.028
Pneumonia	0 (0)	3 (4.2)	5 (7.1)	8 (3.9)	--	0.058	--
Rash	0 (0)	5 (7.0)	3 (4.3)	8 (3.9)	0.059	--	--
Urinary incontinence*	6 (9.1)	0 (0)	1 (1.4)	7 (3.4)	0.011	0.057	--
Blood Glucose Increased	4 (6.1)	0 (0)	1 (1.4)	5 (2.4)	0.051	--	--

Number of patients (%) p value

Safety: Patients with Adverse Events by Treatment Group

System Organ Class	Placebo	400 mg	800 mg
Blood/lymphatics (primarily eosinophilia)	1	5	11
Cardiac	1	6	5
Gastrointestinal	21	20	26
General disorders	10	16	15
Infections/infestations	20	18	19
Metabolism	5	6	7
Musculoskeletal	10	11	13
Nervous system	13	20	14
Psychiatric disorders	23	26	14
Renal/urinary disorders	8	4	8
Respiratory/thoracic	12	17	10
Skin/subcutaneous tissue	8	12	11
Vascular disorders (primarily elevated bp)	1	8	6

Safety Summary

- Overall, MPC-7869 appeared very well tolerated
- Discontinuations due to AEs were comparable between 800 mg BID and placebo
- Adverse events (higher frequency than placebo)
 - transient eosinophilia, mild anemia, bp elevation, lower respiratory infection, rash
- Adverse events (lower frequency than placebo)
 - psychiatric events (p=0.057, placebo vs. 800 mg BID)
 - urinary incontinence

Psychiatric Disorder Adverse Events Mild Patients (MMSE ≥ 20), Safety Population

Psychiatric Disorders (MedDRA)	Placebo (N=50)	400 mg BID (N=40)	800 mg BID (N=50)	Total (N=140)
Total Number of Adverse Events	21	17	10	48
Number of Patients With at Least One Adverse Event (Psychiatric Disorder)	18 (36%)	12 (30%)	7 (14%)	37 (26.4%)
Abnormal behaviour	1 (2%)	0	0	1 (0.7%)
Abnormal dreams	1 (2%)	0	0	1 (0.7%)
Aggression	2 (4%)	0	0	2 (1.4%)
Agitation	3 (6%)	2 (5%)	1 (2%)	6 (4.3%)
Anger	0	1 (2.5%)	0	1 (0.7%)
Anxiety	2 (4%)	2 (5%)	2 (4%)	6 (4.3%)
Apathy	1 (2%)	0	0	1 (0.7%)
Confusional state	2 (4%)	4 (10%)	0	6 (4.3%)
Delusion	0	1 (2.5%)	0	1 (0.7%)
Depression	2 (4%)	1 (2.5%)	4 (8%)	7 (5.0%)
Hallucination	1 (2%)	0	0	1 (0.7%)
Hallucination, visual	1 (2%)	1 (2.5%)	0	2 (1.4%)
Insomnia	1 (2%)	0	1 (2%)	2 (1.4%)
Libido increased	1 (2%)	1 (2.5%)	0	2 (1.4%)
Mood altered	0	0	1 (2%)	1 (0.7%)
Mood swings	0	1 (2.5%)	0	1 (0.7%)
Nightmare	1 (2%)	0	1 (2%)	2 (1.4%)
Paranoia	0	1 (2.5%)	0	1 (0.7%)
Psychotic disorder	0	1 (2.5%)	0	1 (0.7%)
Sleep disorder	1 (2%)	1 (2.5%)	0	2 (1.4%)

p = 0.020 (Placebo vs. 800 mg BID)

Time to Psychiatric Event by Treatment Group (*Mild Patients)

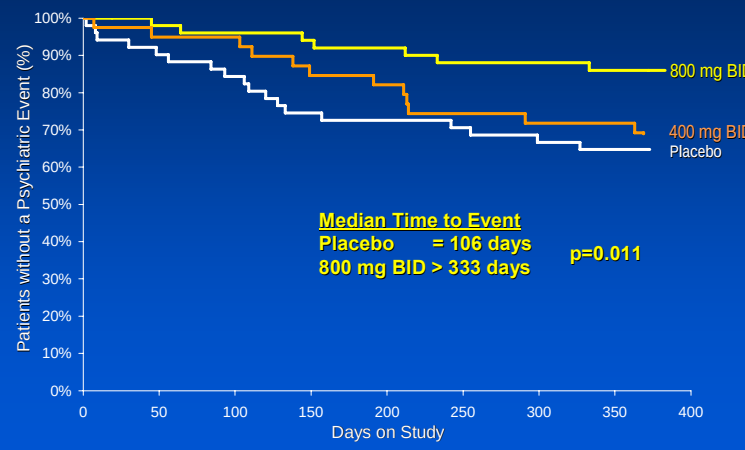
100%
90%
80%
70%
60%
50%
40%
30%
20%
10%
0%
Patients without a Psychiatric Event (%)
0
50
100
150
200
250
300
350
400
Days on Study
800 mg BID
400 mg BID
Placebo
Median Time to Event
Placebo = 106 days
800 mg BID > 333 days
p=0.011

*MMSE ≥ 20 at Baseline

Conclusions

- MPC-7869 has an attractive therapeutic and safety profile in patients with mild AD treated for 1 year, the vast majority of whom were already on standard of care therapy (acetylcholinesterase inhibitors)
- In addition to the reported[1] significant benefit observed in patients with mild AD in activities of daily living (p=0.033), global function (p=0.042) and a positive trend in cognition, this analysis revealed a significant reduction (p=0.020) in the number of and a delay in time to psychiatric events (p=0.011)
 - The most common psychiatric events reported in the placebo group were agitation, aggression, confusional state and depression
 - No significant effect was seen in the NPI (Neuropsychiatric Inventory) exploratory endpoint
- These results are consistent with the hypothesis that treatment with MPC-7869 may delay progression of AD
- Confirmatory Phase 3 Studies are ongoing

Reference:
[1]Black, SE et al. 2006. Neurology 66(5), Supplement 2, pA347

PDF copies of this poster are available at www.myriad.com